Predictive Value of Mini-BESTest Subdomains for Future Falls in Patients with Parkinson's Disease

M. Brogan, DPT[1]; M. Kilgore, DPT[1]; S. Nithiananda, DPT[1]; H. Williford, DPT[1]
O. Shafer[2]; J.L. McKay PhD, MSCR[3]
[1] Emory University Department of Physical Therapy, Rollins School of Public Health, Emory University, [3]Coulter Department of Biomedical Engineering Emory University and Georgia Tech

Supported by NIH K25 HD086276

Introduction

Patients with Parkinson's disease (PD) are at an increased risk for falls. The Mini-BESTest is a clinical measure designed to challenge distinct balance domains. An improved understanding of PD-specific balance and fall mechanisms is needed to provide optimal Physical Therapy treatment. The goal of this study is to determine which subdomain of the Mini-BESTest best predicts future falls in PD patients.

Hypothesis

Mini-BESTest Subdomain 2 - Reactive Postural Control will be most predictive of future falls when compared to other subdomains in patients with Parkinson's disease.

Study Design

- Prospective observational study embedded within a 3-year observational study of fall risk
- 48 patients with Parkinson's disease (n=47 included in analysis)
- Outcome Measures:
 - Functional Assessment: Mini Balance Evaluation Systems Test (Mini-BEST)
 - Cognitive Assessment: Montreal Cognitive Assessment (MoCA)
 - Movement Disorders Society Unified Parkinson's Disease Rating Scale (MDS-UPDRS)
 - Freezing of Gait Questionnaire (FOG-Q)

Data Collection

1. Informed consent and study enrollment
2. Collection of demographic information
3. Cognitive assessments
4. Functional assessments
5. MDS-UPDRS-III (Rated by SA Factor, DO)
6. Preparation and collection of longitudinal study measurements (electromyography + dynamic posturography)
7. Monthly fall report collection for 12 months
8. Data analysis performed by O Shafer

Results

1. We assessed n=47 patients with PD in the 12-hours off-medication state.

Characteristic	Total (n=47)
Sex	
Male, n (%)	30 (64)
Female, n (%)	17 (36)
Freezer	
Yes, n (%)	39 (83)
No, n (%)	8 (17)
Age today, (y) σ	66 ± 7
PD Duration, (y) σ	9 ± 6
MDS-UPDRS-III	33.5 ± 13.5
MoCA	26.9 ± 2.3
Mini-BESTest	21.6 ± 4.3
LEDD, median (range)	700 (0-2240)

Reported as mean ± standard deviation (σ) unless otherwise noted.

2. Submaximal performance on Mini-BESTest Subdomains 1-3 were associated with increased fall rate, with Subdomain 2 having the strongest association (Rate Ratio: 31.3, 95% CI: 15.8-72.8).

	Not Perfect Score	Perfect Score	Total	Rate Ratio	95% CI
Subdomain 1: Anticipatory Postural Adjustments	n=41	n=6	n=47	**5.4***	3.5-8.7
Number of Falls	685	22	707		
Person-Months	323	56	379		
Subdomain 2: Reactive Postural Control	n=33	n=14	n=47	**31.3***	15.8-72.8
Number of Falls	699	8	707		
Person-Months	279	100	379		
Subdomain 3: Sensory Orientation	n=24	n=23	n=47	**1.3***	1.1-1.5
Number of Falls	387	320	707		
Person-Months	184	195	379		

3. Submaximal performance on Mini-BESTest Subdomain 4 was associated with decreased fall rate.

	Not Perfect Score	Perfect Score	Total	Rate Ratio	95% CI
Subdomain 4: Dynamic Gait	n=39	n=8	n=47	**0.4***	.37-.43
Number of Falls	391	316	707		
Person-Months	292	87	379		

Discussion

- The significantly increased rate of falls in patients with submaximal Subdomain 2 scores suggests that a decreased ability to respond to external balance challenges is predictive of future falls.
- Submaximal performance on Subdomain 4 was found to be protective against future falls. This may be due to an increase in risk-taking behaviors during gait and an increased exposure to fall risk.

Conclusion

Early identification of fall risk and appropriate physical therapy treatment are crucial to the safety and quality of life of individuals with PD. This study identified a strong association between submaximal score on Subdomain 2 of the Mini-BESTest and future falls in patients with PD. This quick, easy-to-score balance exam appears to specifically predict an increased future rate of falls in this population.

References

1. Behling, C., Jacobs, K., Rift, E., Su, S., Volden, M., Kiang, C., & McKay, J. L. (2018). Reactive Postural Control on the Mini-BEST is differentially associated with Fall History in Parkinson's Disease. 1.
2. Bloem, B. R., Grimbergen, Y. A., Cramer, M., Willemsen, M., & Zwinderman, A. H. (2001). Prospective assessment of falls in Parkinson's disease. Journal of Neurology, 248(11), 950–958.
3. Duncan, R. P., Leddy, A. L., Cavanaugh, J. T., Dibble, L. E., Ellis, T. D., Ford, M. P., … Earhart, G. M. (2012). Accuracy of Fall Prediction in Parkinson Disease: Six-Month and 12-Month Prospective Analyses. Parkinson's Disease. https://doi.org/10.1155/2012/237673
4. Duncan, R. P., Leddy, A. L., Cavanaugh, J. T., Dibble, L. E., Ellis, T. D., Ford, M. P., … Earhart, G. M. (2013). Comparative utility of the BESTest, mini-BESTest, and brief-BESTest for predicting falls in individuals with Parkinson disease: a cohort study. Physical Therapy, 93(4), 542–550. https://doi.org/10.2522/ptj.20120302
5. Hoops, S., Nazem, S., Siderowf, A. D., Duda, J. E., Xie, S. X., Stern, M. B., & Weintraub, D. (2009). Validity of the MoCA and MMSE in the detection of MCI and dementia in Parkinson disease. Neurology, 73(21), 1738–1745. https://doi.org/10.1212/WNL.0b013e3181c34b47
6. Leddy, A. L., Crowner, B. E., & Earhart, G. M. (2011). Utility of the Mini-BESTest, BESTest, and BESTest sections for balance assessments in individuals with Parkinson disease. Journal of Neurologic Physical Therapy: JNPT, 35(2), 90–97. https://doi.org/10.1097/NPT.0b013e31821a620c
7. McKay, J. L., Lang, K. C., Ting, L. H., & Hackney, M. E. (2018). Impaired set shifting is associated with previous falls in individuals with and without Parkinson's disease. Gait & Posture, 62, 220–226. https://doi.org/10.1016/j.gaitpost.2018.02.027
8. Paul, S. S., Canning, C. G., Sherrington, C., Lord, S. R., Close, J. C. T., & Fung, V. S. C. (2013). Three simple clinical tests to accurately predict falls in people with Parkinson's disease. Movement Disorders: Official Journal of the Movement Disorder Society, 28(5), 655–662. https://doi.org/10.1002/mds.25404
9. van der Marck, M. A., Klok, M. Ph. C., Okun, M. S., Giladi, N., Munneke, M., & Bloem, B. R. (2014). Consensus-based clinical practice recommendations for the examination and management of falls in patients with Parkinson's disease. Parkinsonism & Related Disorders, 20(4), 360–369. https://doi.org/10.1016/j.parkreldis.2013.10.030
10. Wood, B. H., Bilclough, J. A., Bowron, A., & Walker, R. W. (2002). Incidence and prediction of falls in Parkinson's disease: a prospective multidisciplinary study. Journal of Neurology, Neurosurgery & Psychiatry, 72(6), 721–725. https://doi.org/10.1136/jnnp.72.6.721